Present and Future Health and Economic Burden of Controlled and Uncontrolled Gout in Patients with Chronic Kidney Disease in the United States

POS0509

Joshua Card-Gowers (a), Laura Webber (a), Lise Retat (a), Marek Piotrowski (a), Brian LaMoreaux (b), Brad Marder (b), Ada Kumar (b)

Introduction

- The prevalence of comorbid gout and chronic kidney disease (CKD) is high in the US, posing a large burden to patients and the healthcare system.[1]
- Current gout management in the US is sub-optimal, with many patients remaining untreated, undertreated, or uncontrolled.[2]
- Understanding the current and future burden of coincident gout and CKD is important for adequate health and economic resource planning.

Objective

- Quantify and compare the current, and project the future, health and economic burden of both controlled and uncontrolled gout in the US CKD population between 2023 and 2035.

Methods

A validated microsimulation model was used for this study.[3]

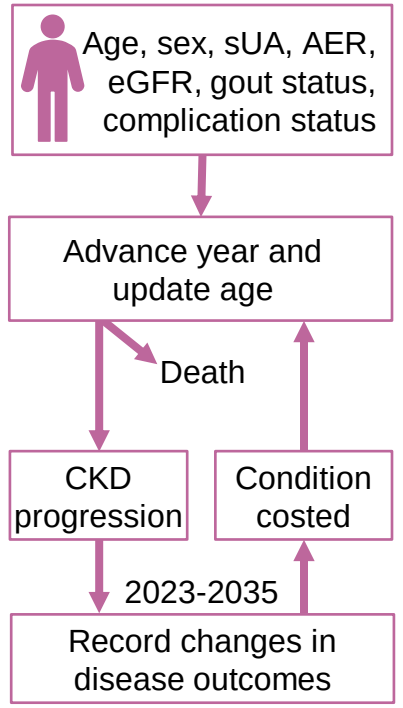
Age, sex, sUA, AER, eGFR, gout status, complication status
Advance year and update age
Death
CKD progression
Condition costed
2023-2035
Record changes in disease outcomes

AER, albumin excretion rate; eGFR, estimated glomerular filtration rate; NHANES, National Health and Nutrition Examination Survey; sUA, serum uric acid

1. The US population was reproduced, virtually, at an individual level.
2. Individuals were assigned health characteristics based on databases, including the NHANES, and the literature.
3. Controlled and uncontrolled gout costs, CKD progression rates, tophi, and flare probabilities were drawn from published literature.
4. Uncontrolled gout was defined as SU >6 mg/dL and the presence of gout symptoms (≥2 flares a year or tophi).
5. The microsimulation was run between 2023 and 2035, allowing individuals' health states to change each year

Epidemiological Results

- By 2035, 17.4% of CKD patients are projected to have gout, of which, 28% will be classified as uncontrolled.
- This study projects a **2.2% average annual growth** in the prevalence of comorbid gout and CKD in the US between 2023 and 2035, from **8.2 million** (2.4% of the US population) to **10.5 million** (2.9% of the US population).
- The number of people living with comorbid uncontrolled gout and CKD is, on average, projected to increase by 54,000 each year between 2023 and 2035.
- The number of people living with gout and advanced (stages 3-5) CKD is projected to grow by **37.5%** by 2035, of which **29.2%** (**4.77 million** patients) are projected to have uncontrolled gout (Figure 1).
- The number of gout flares each year in the CKD population is projected to increase by 22.2%, from **5.03 million** in 2023 to **6.15 million** in 2035 (Figure 2).
- A projected 3.2 million US CKD patients will have tophaceous gout by 2035, a 21.0% increase from 2023 (Figure 3).

Figure 2. Number of gout flares in the CKD population

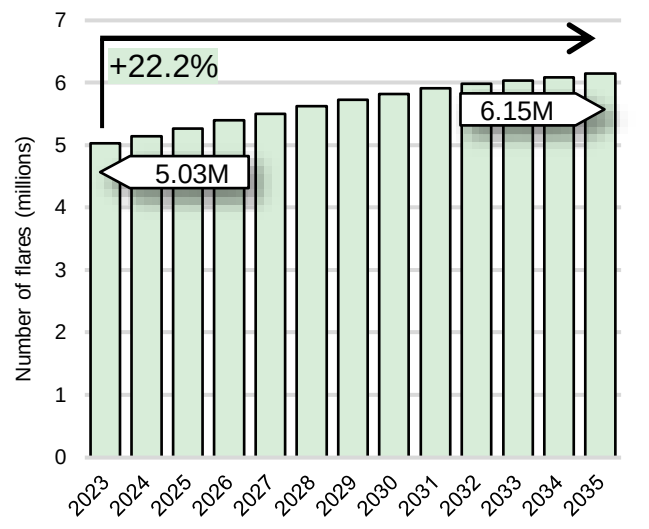
Number of flares (millions)
0
1
2
3
4
5
6
7
+22.2%
5.03M
6.15M
2023 2024 2025 2026 2027 2028 2029 2030 2031 2032 2033 2034 2035

Figure 3. Tophaceous gout cases in the CKD population

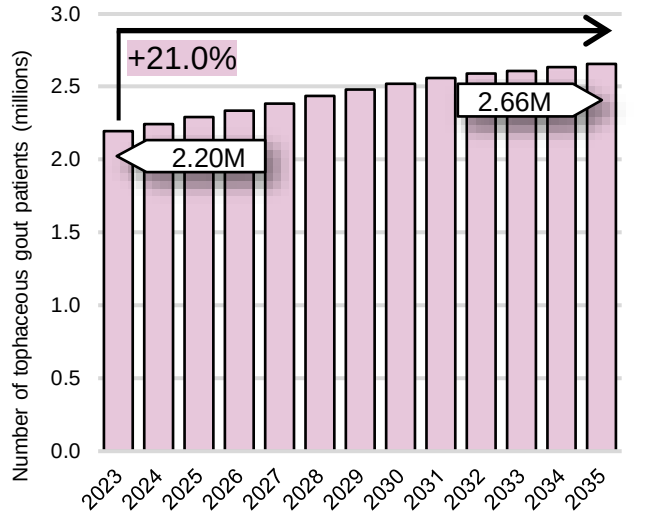
Number of tophaceous gout patients (millions)
0.0
0.5
1.0
1.5
2.0
2.5
3.0
+21.0%
2.20M
2.66M
2023 2024 2025 2026 2027 2028 2029 2030 2031 2032 2033 2034 2035

Figure 1. Prevalence of comorbid gout and CKD in the US

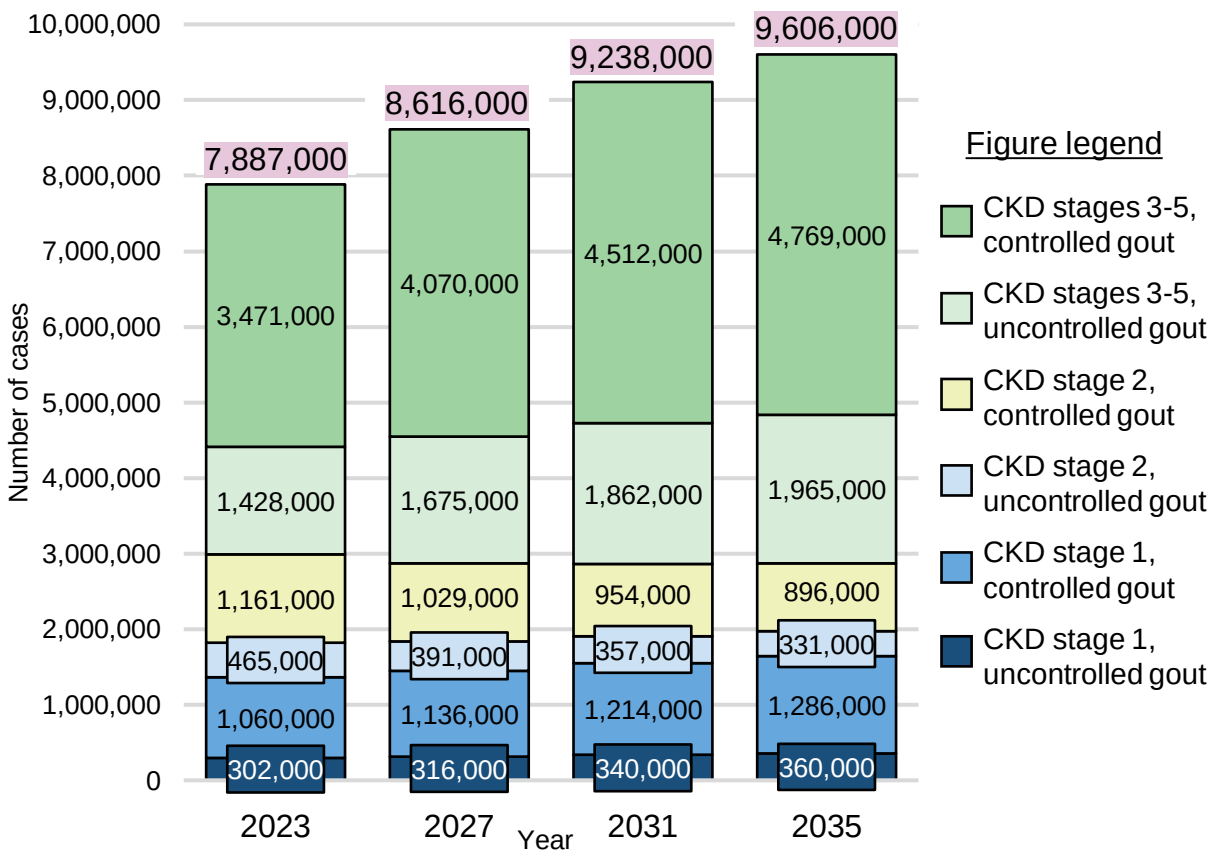
Number of cases
0
1,000,000
2,000,000
3,000,000
4,000,000
5,000,000
6,000,000
7,000,000
8,000,000
9,000,000
10,000,000
7,887,000
8,616,000
9,238,000
9,606,000
3,471,000
4,070,000
4,512,000
4,769,000
1,428,000
1,675,000
1,862,000
1,965,000
1,161,000
1,029,000
954,000
896,000
465,000
391,000
357,000
331,000
1,060,000
1,136,000
1,214,000
1,286,000
302,000
316,000
340,000
360,000
2023
2027
2031
2035
Year
Figure legend
CKD stages 3-5, controlled gout
CKD stages 3-5, uncontrolled gout
CKD stage 2, controlled gout
CKD stage 2, uncontrolled gout
CKD stage 1, controlled gout
CKD stage 1, uncontrolled gout

Economic Results

- The combined direct and indirect costs of gout in CKD patients are projected to increase from **$39.1B** in 2023 to **$50.4B** by 2035; equivalent to **$588.2B** cumulatively over the next 12 years, of of which **$228.6B** is attributable to uncontrolled gout (Figure 4).
- If all gout in CKD was controlled, the US could save **$86.2B** through 2035.
- A projected **51 million** working days are to be lost due to uncontrolled gout in the US, between 2023 and 2035; an average of **3.78 million** working days lost each year.

Figure 4. Costs of gout in patients with CKD

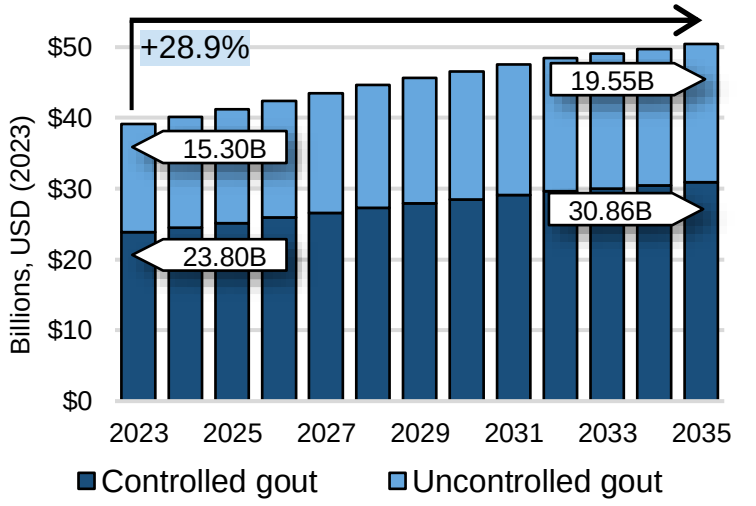
Billions, USD (2023)
$0
$10
$20
$30
$40
$50
+28.9%
15.30B
23.80B
19.55B
30.86B
2023 2025 2027 2029 2031 2033 2035
Controlled gout
Uncontrolled gout

Conclusions

- The number of people living with comorbid gout and CKD is projected to markedly increase in coming years, with associated direct and indirect costs.
- Uncontrolled gout is projected to heavily contribute to the burden of comorbid gout and CKD.
- Results suggest that interventions to reduce serum urate and control gout could substantially lessen the burden of disease for patients and the US health system.

References: [1] Krishnan E, et al. *PloS One* 2012;7:e50046; [2] Edwards NL, et al. ACR Open Rheumatol 2020;2:180-7. [3] Tangri N, et al. Adv Ther 2022;1-17. **Affiliations**: a) HealthLumen, London, UK; b) Horizon Therapeutics, Deerfield, Illinois, US. **Disclosures**: JCG, LW, LR, and MP are employees of HealthLumen, who was contracted by Horizon for forecasting expertise. BL, BM, and AK are employees and stockholders in Horizon Therapeutics. Presented at EULAR 2023 (May 31 – June 3; Milan, Italy). This research was funded by Horizon Therapeutics, PLC.